Conquering Disease
With Targeted
Protein Degradation

Development of AchillesTAG degradation systems and their application to control CAR-T activity

C4 Therapeutics team

490 Arsenal Way, Watertown, MA 02472, USA

BACKGROUND

In recent years, several chemical genetic approaches have emerged that enable the rapid and acute control of target protein homeostasis. These technologies include the auxin-inducible degron (AID)[1], dTAG system[2], HaloTAG PROTACS[3,4], small-molecule-assisted shutoff (SMASh)[5], cryptic degrons[6], and the Shield system[7]. These sophisticated technologies have elevated the understanding of target biology in both *in vitro* and *in vivo* settings and exemplified the power of rapid target protein modulation. With these technologies as an inspiration, we aimed to develop additional small molecular degradation tags that would allow for the pharmacologic control of protein homeostasis via heterobifunctional molecule mediated target degradation. We developed two non-overlapping aTAG systems that provide the flexibility in choice of aTAG properties and the opportunity to multiplex aTAG systems within the same context to further explore complex biology. Each aTAG system comes with a suite of degrader molecules with a range of *in vivo* properties that allow for optionality to meet the needs of a given *in vivo* application. We applied these aTAG systems as a solution to the increasing demand of mechanisms to control Chimeric Antigen Receptor T cell (CAR-T) activity. We show that pharmacologic control of a chimeric antigen receptor (CAR) via targeted protein degradation (mediated by an aTAG) provides a cell-sparing, reversible, and tunable approach.

AchillesTAG DEGRADATION SYSTEMS

Engineering novel degradation tags: aTAG

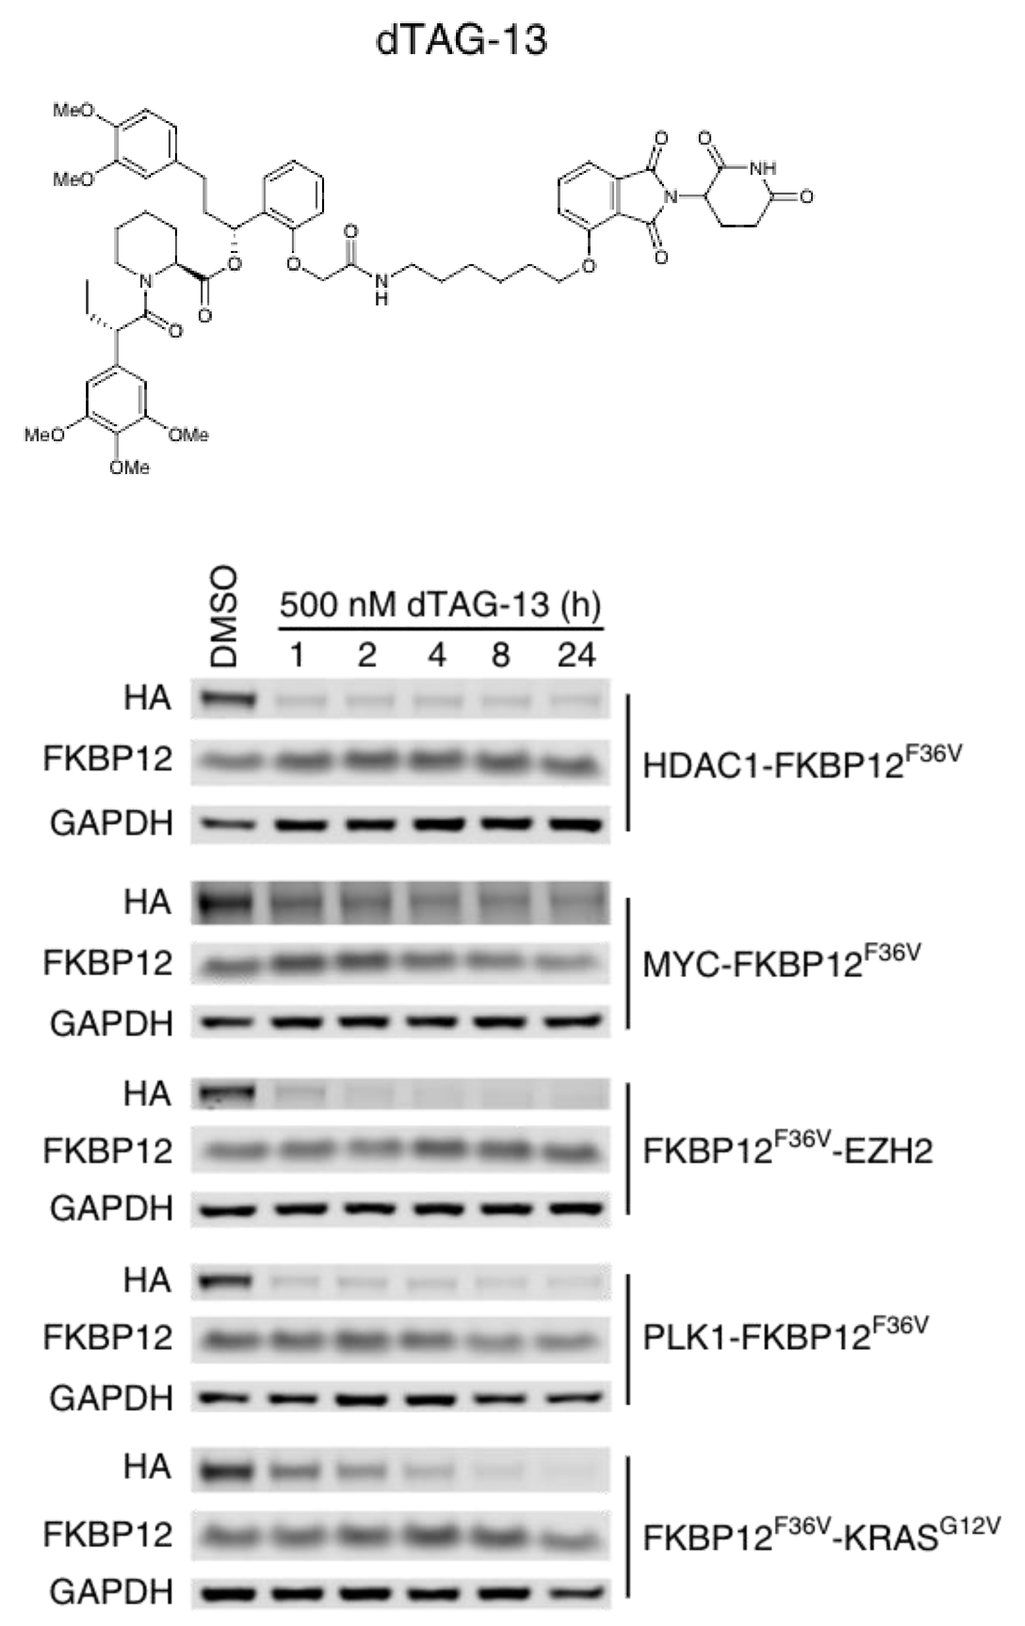
dTAG-13
DMSO
500 nM dTAG-13 (h)
1 2 4 8 24
HA
FKBP12
GAPDH
HDAC1-FKBP12F36V
MYC-FKBP12F36V
FKBP12F36V-EZH2
PLK1-FKBP12F36V
FKBP12F36V-KRASG12V

Nabet et al., 2018

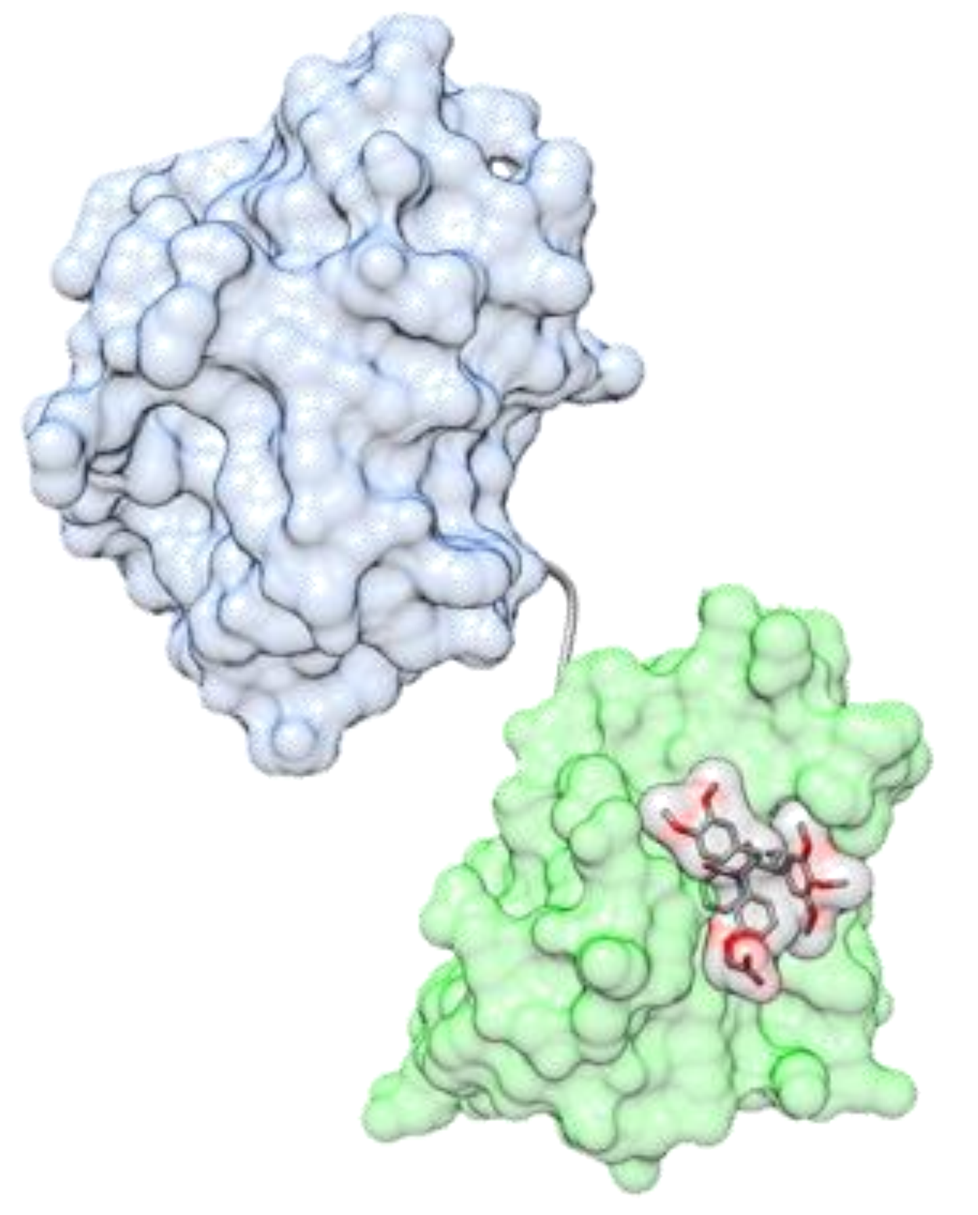

Introduce an *Achilles'* heel
into the protein of interest
rendering the overall chimera degradable
in a cereblon-dependent fashion

Exemplified with FKBP12* dTAG

AchillesTAG = aTAG

Development of novel aTAG systems

DESIGN CRITERIA

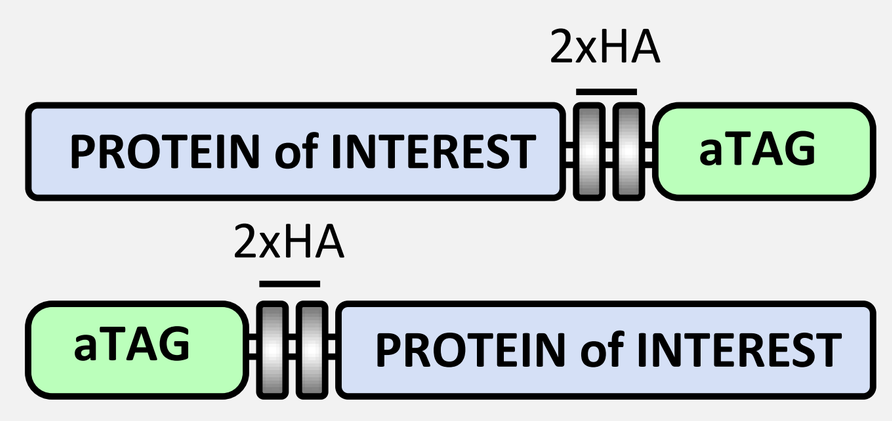
2xHA
PROTEIN of INTEREST
aTAG
2xHA
aTAG
PROTEIN of INTEREST

DEGRADATION DOMAIN
Small (<20kDa)
Readily 'ligandable'

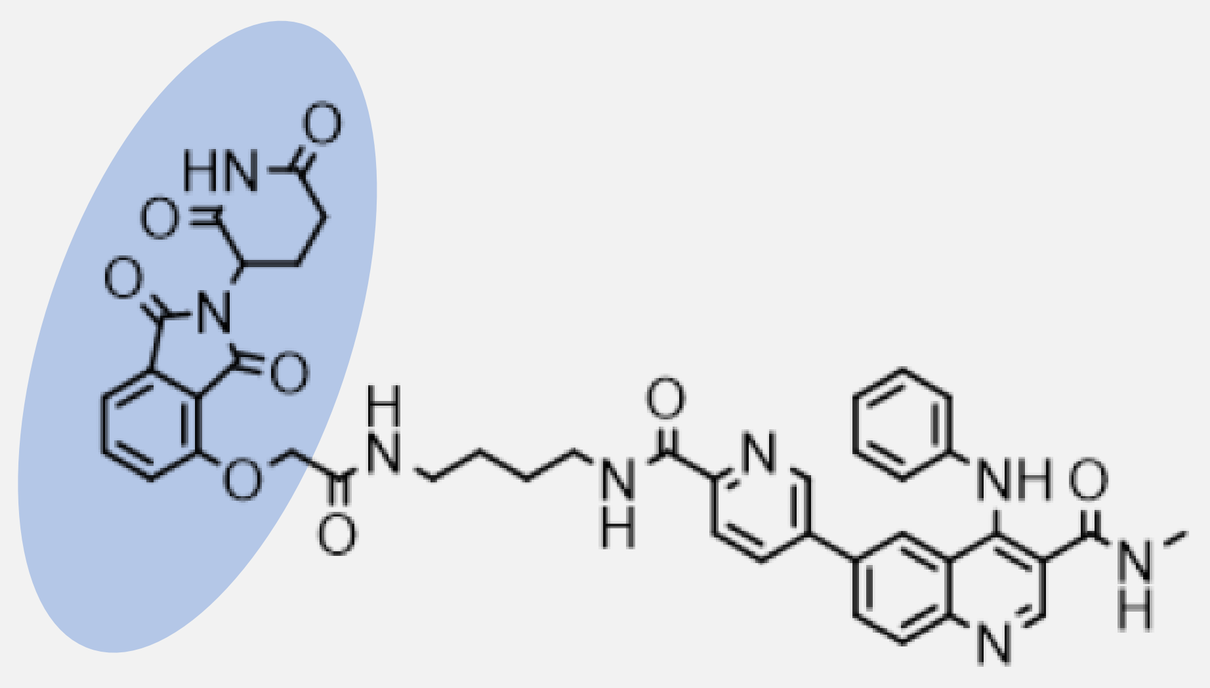

DEGRADER MOLECULES
In vitro potency (DC_{50} < 10nM)
Enabling *in vivo properties*
Clean off target profile

aTAG FUNCTIONAL SCREENING

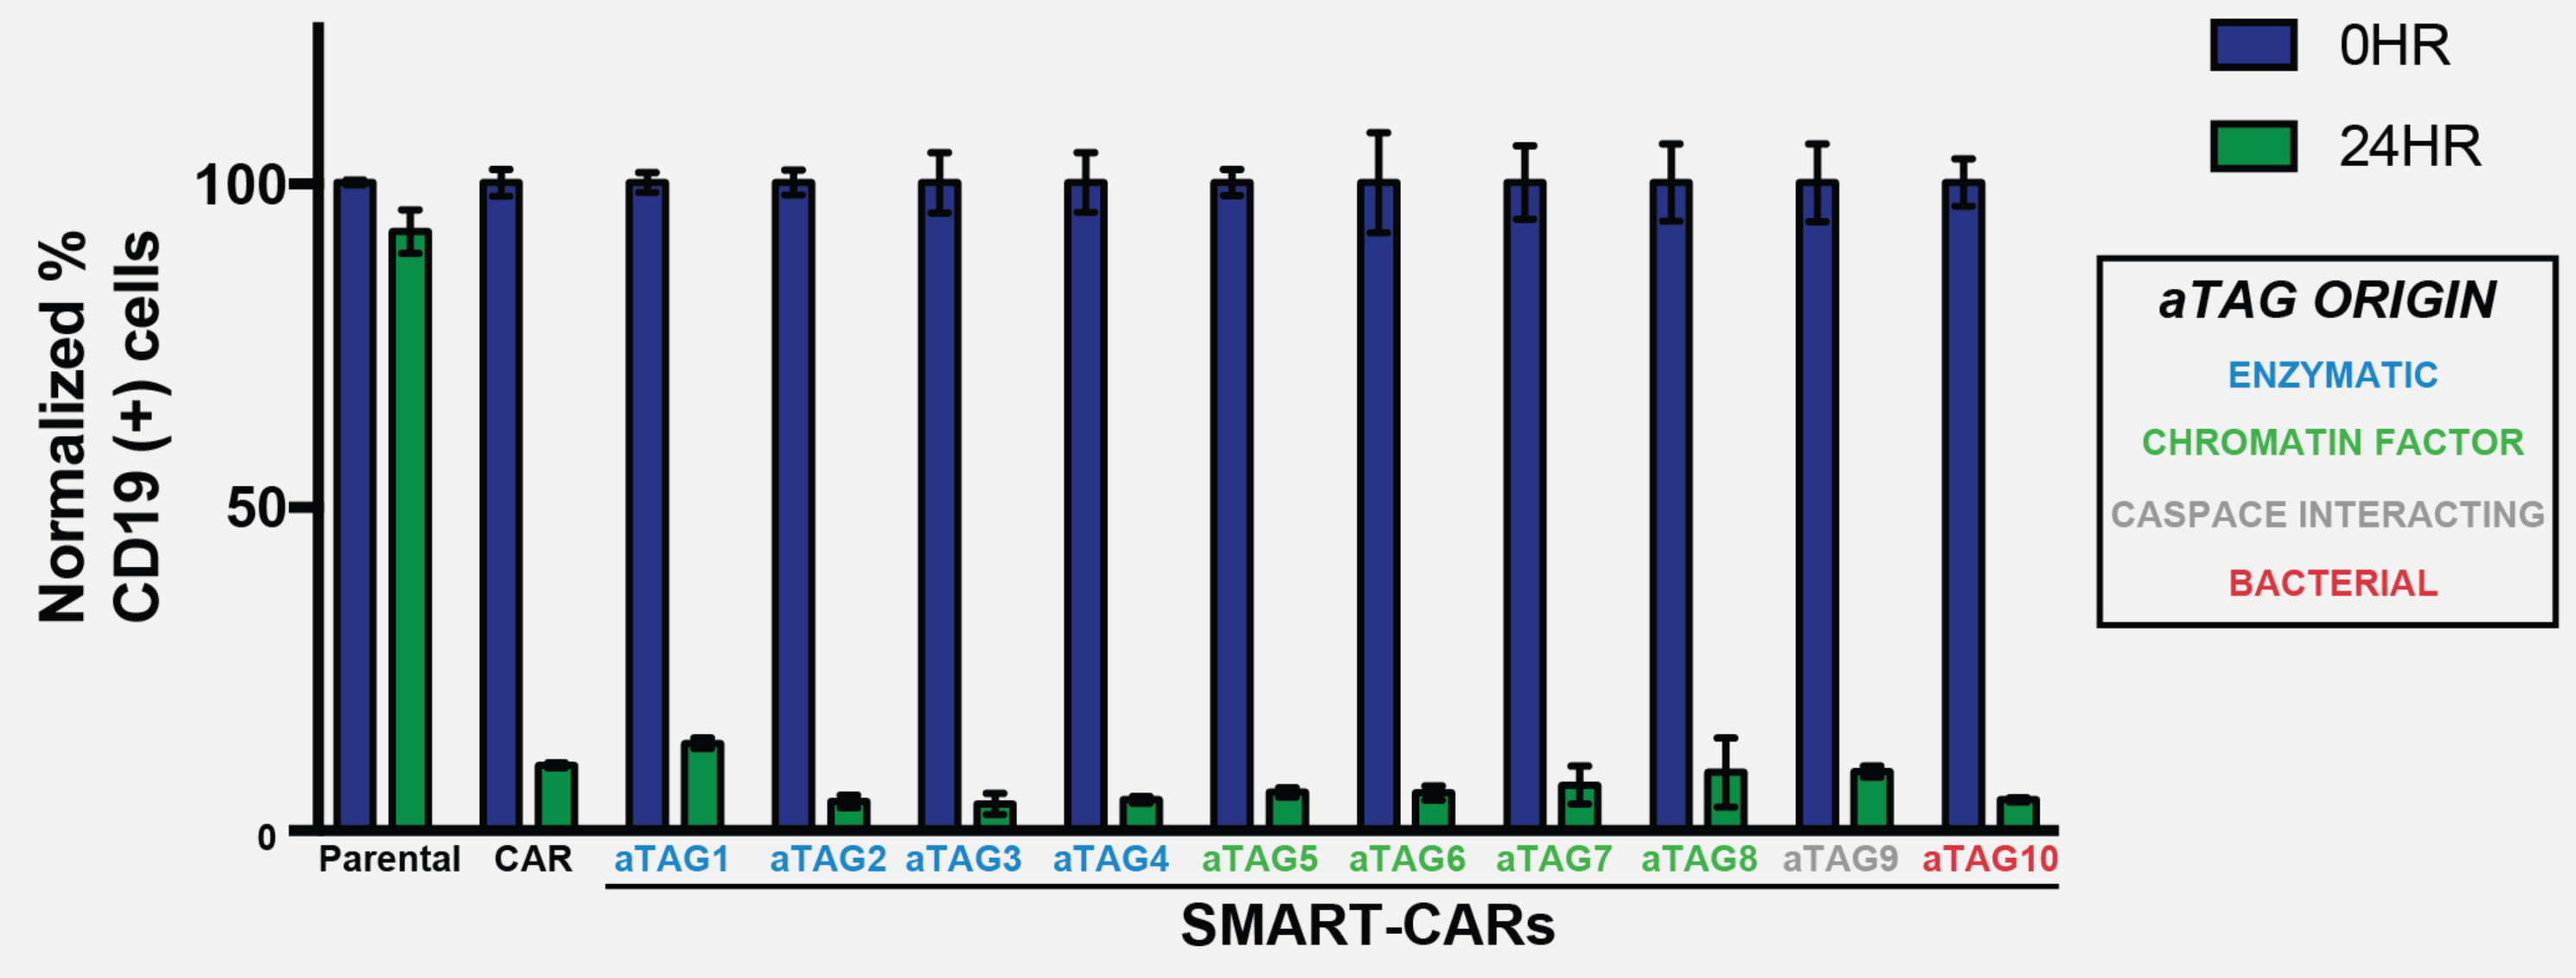
Normalized % CD19 (+) cells
100
50
0
0HR
24HR
Parental
CAR
aTAG1
aTAG2
aTAG3
aTAG4
aTAG5
aTAG6
aTAG7
aTAG8
aTAG9
aTAG10
SMART-CARs
aTAG ORIGIN
ENZYMATIC
CHROMATIN FACTOR
CASPACE INTERACTING
BACTERIAL

Two high fidelity aTAG systems

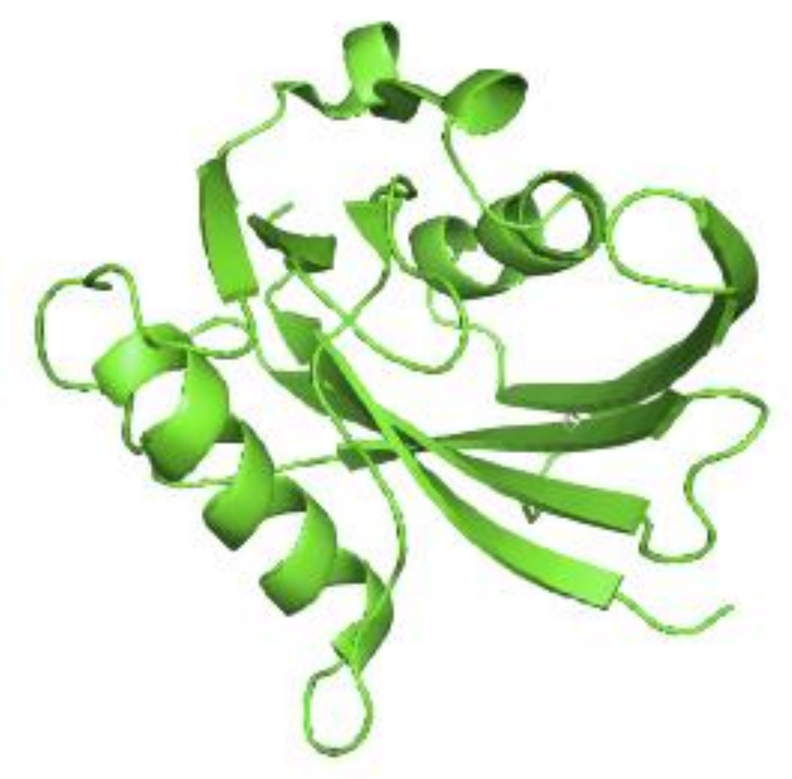

aTAG1 BASED on MTH1 (NUDT1)
Tag size = 17 kDa
Physiologic role: Hydrolysis of 8-oxo-dGTP
NUDT1$^{-/-}$ mice show no phenotype
Acute inhibition or degradation shows no cellular phenotype

CFT-2139

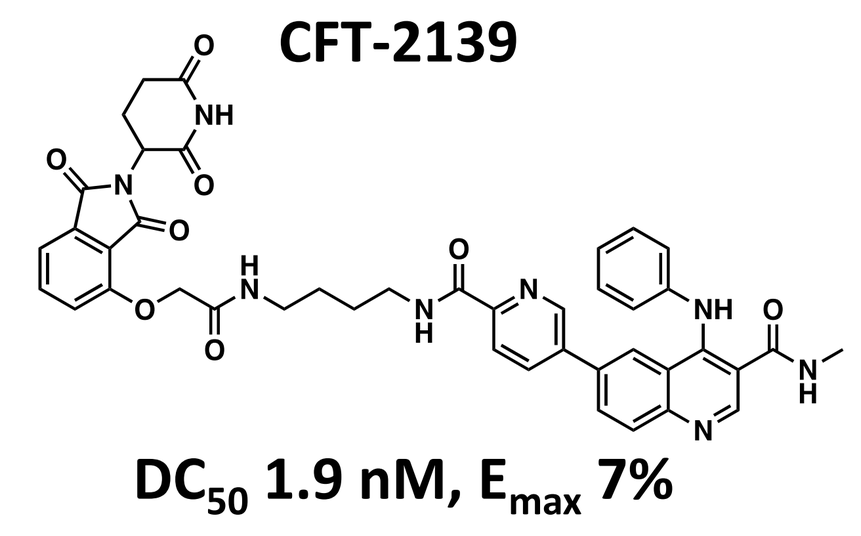

DC_{50} 1.9 nM, E_{max} 7%

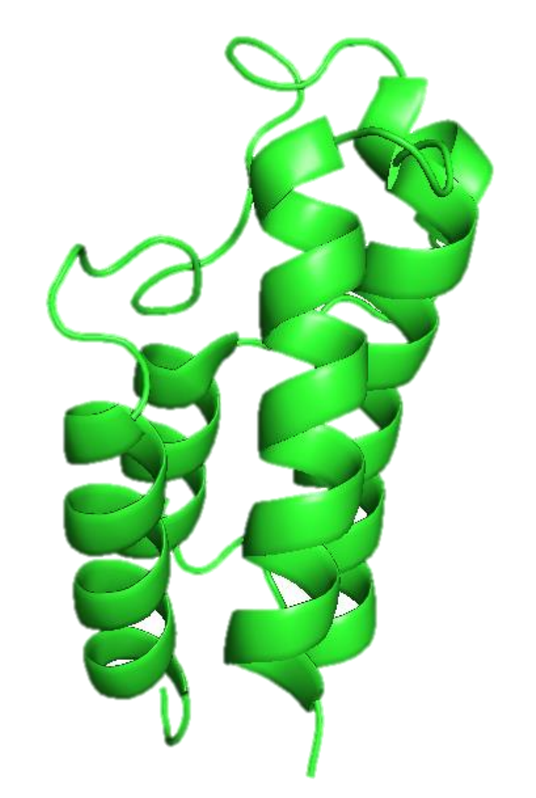

aTAG2 BASED on BRD9
Tag size = 12.5 kDa
Physiologic role: ncBAF complex
Conditional dependency in some AML cells

CFT-6969

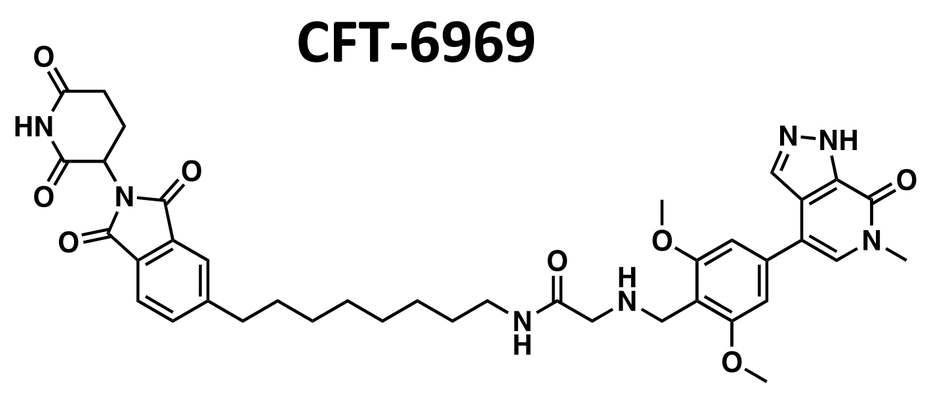

DC_{50} 0.3 nM, E_{max} 2%

aTAGs APPLIED TO CAR-T: SMART-CAR

SMART-CAR concept

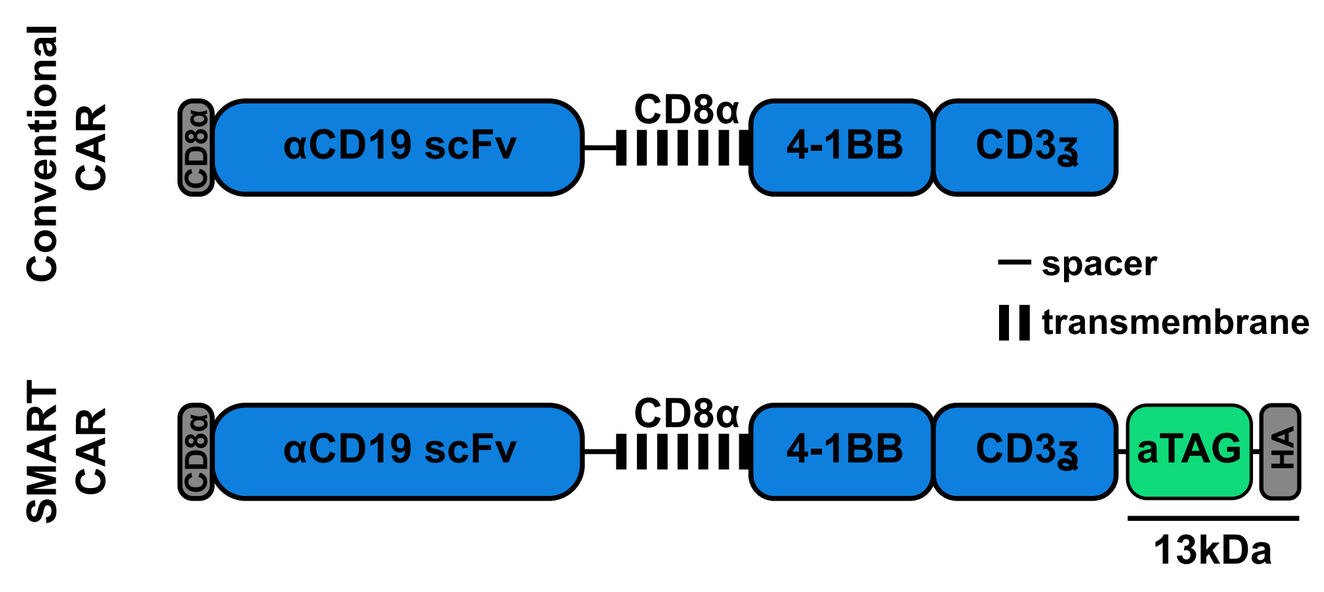
Conventional CAR
SMART CAR
CD8α
αCD19 scFv
CD8α
4-1BB
CD3ζ
aTAG
HA
spacer
transmembrane
13kDa

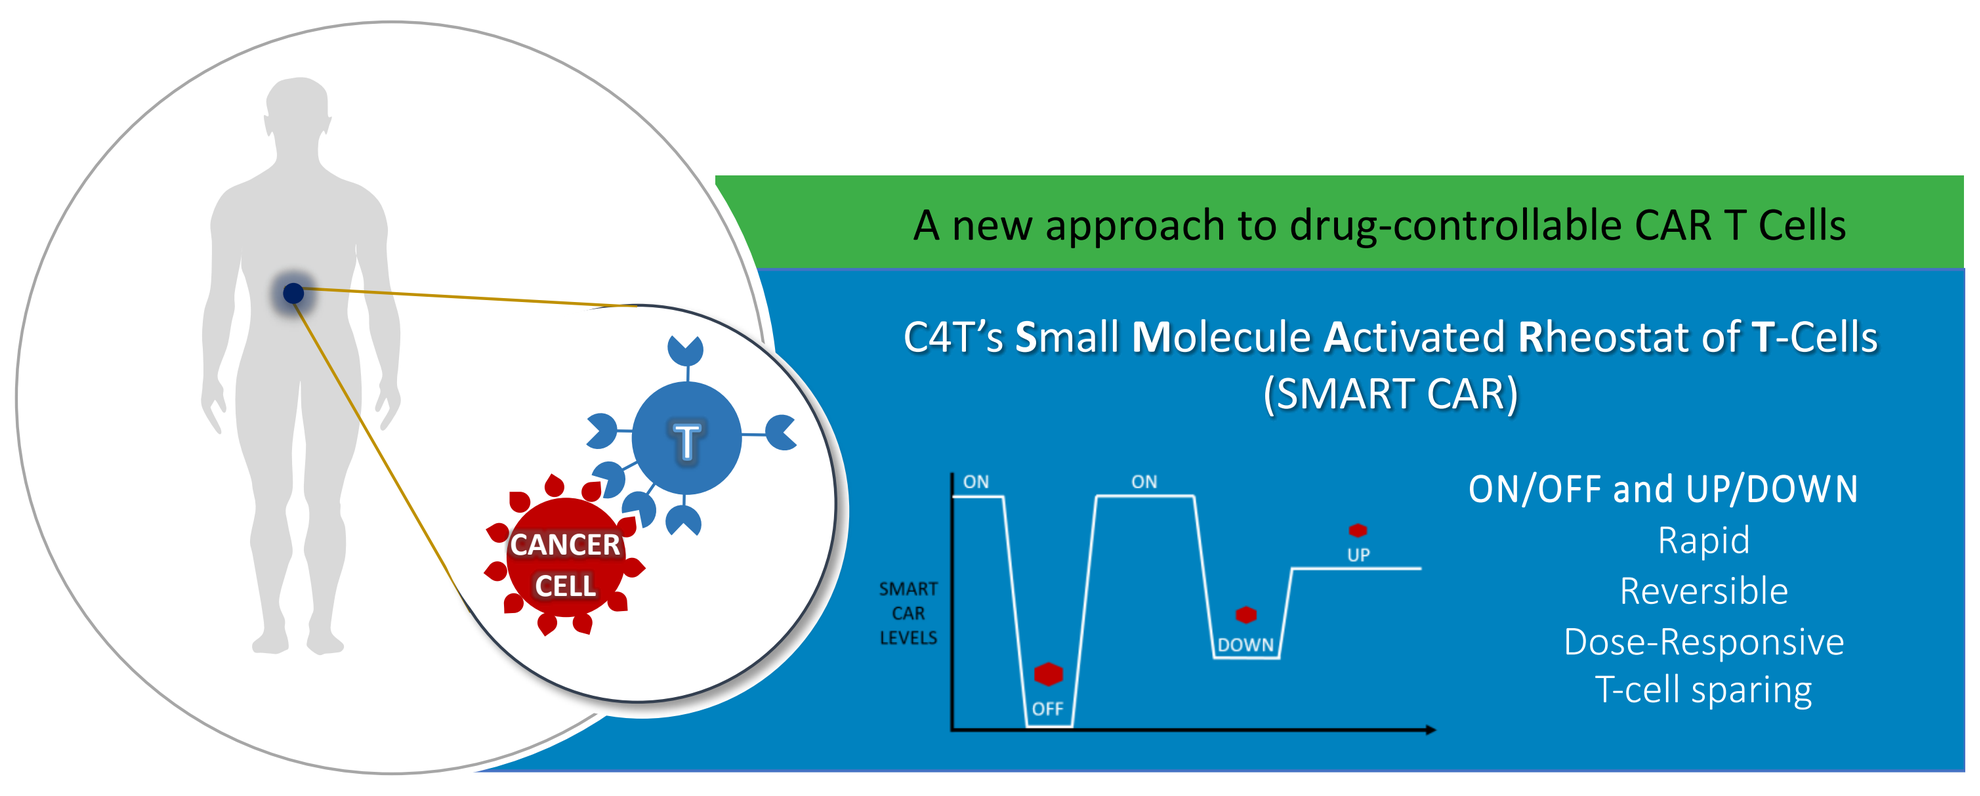
A new approach to drug-controllable CAR T Cells
C4T's Small Molecule Activated Rheostat of T-Cells (SMART CAR)
CANCER CELL
T
SMART CAR LEVELS
ON
OFF
ON
DOWN
UP
ON/OFF and UP/DOWN
Rapid
Reversible
Dose-Responsive
T-cell sparing

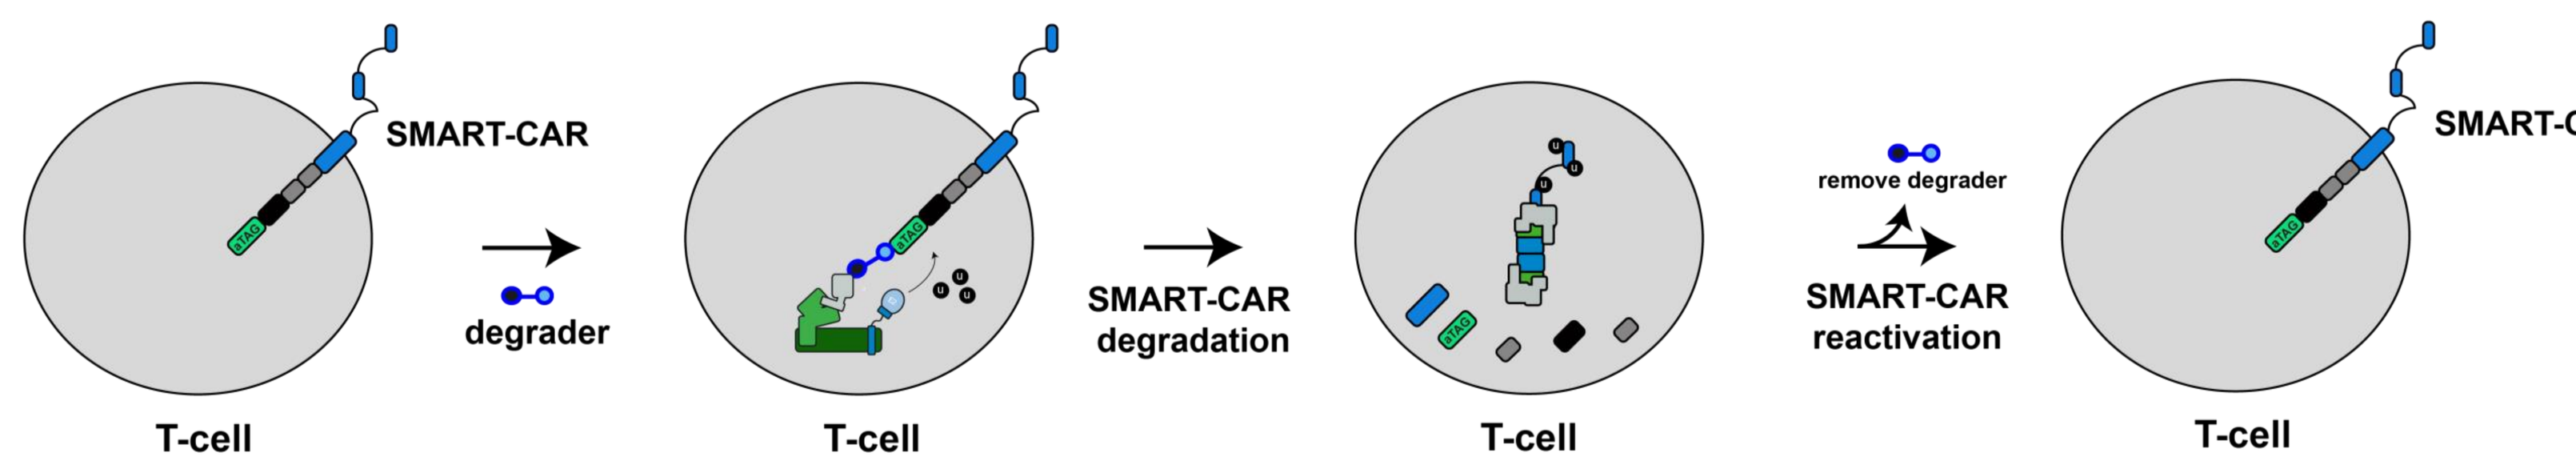
SMART-CAR
degrader
T-cell
SMART-CAR degradation
T-cell
remove degrader
SMART-CAR reactivation
T-cell
SMART-CAR
T-cell

Degradation control

Targeted SMART-CAR degradation

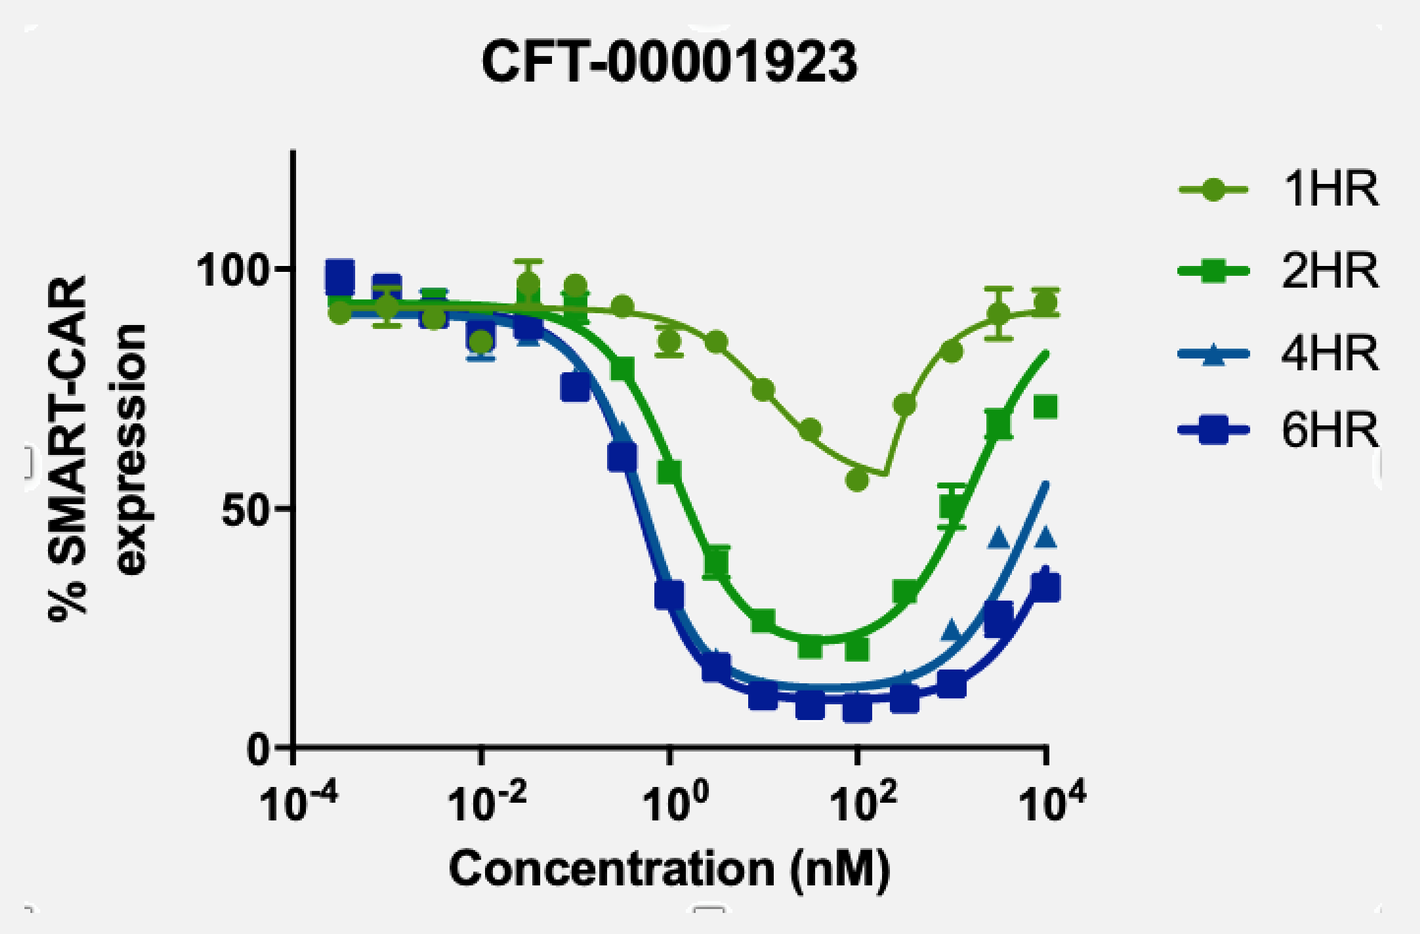
CFT-00001923
% SMART-CAR expression
1HR
2HR
4HR
6HR
Concentration (nM)

Selective degradation

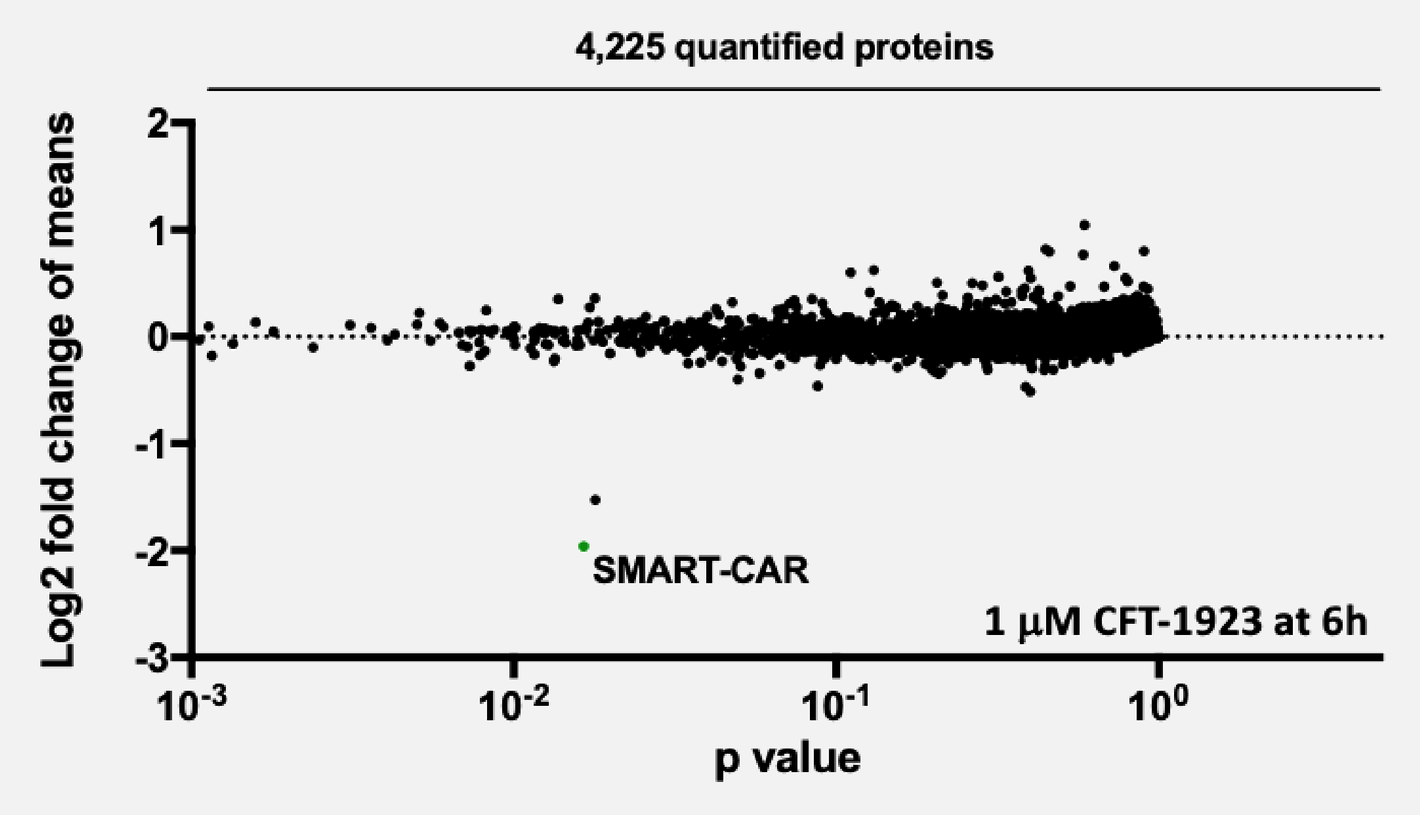
4,225 quantified proteins
Log2 fold change of means
SMART-CAR
1 µM CFT-1923 at 6h
p value

Chemical control of target tumor cell killing

Dose and kinetic control

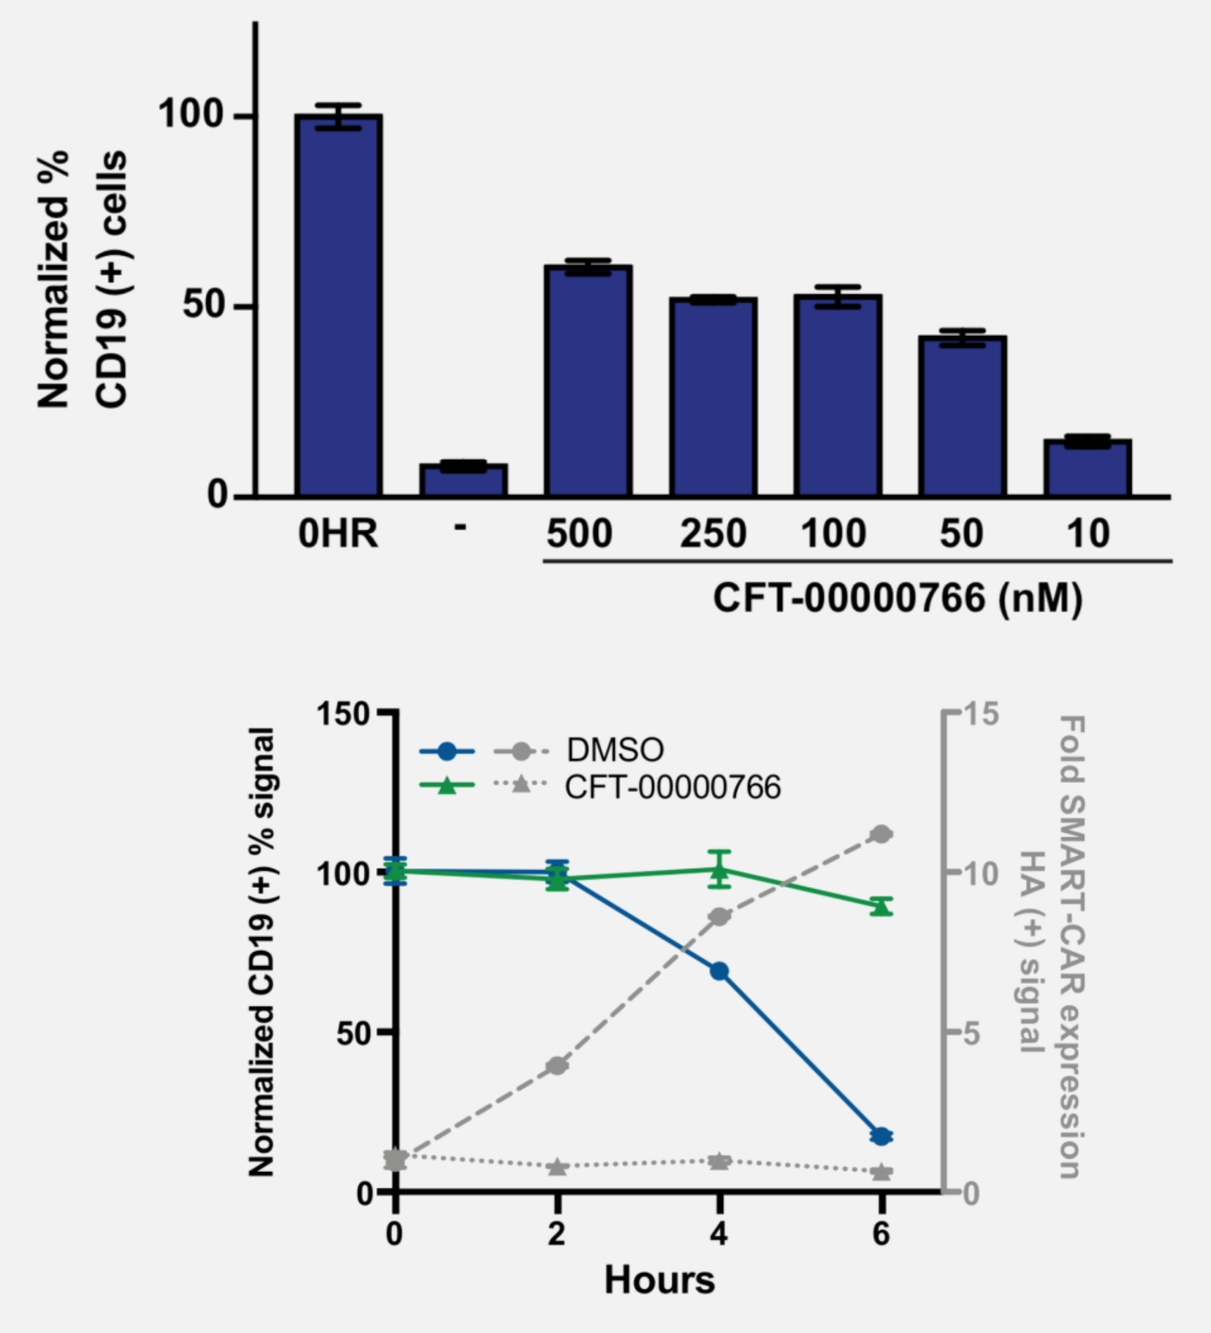
Normalized % CD19 (+) cells
0HR
500
250
100
50
10
CFT-00000766 (nM)
Normalized CD19 (+) % signal
DMSO
CFT-00000766
Fold SMART-CAR expression HA (+) signal
Hours

Rheostat function

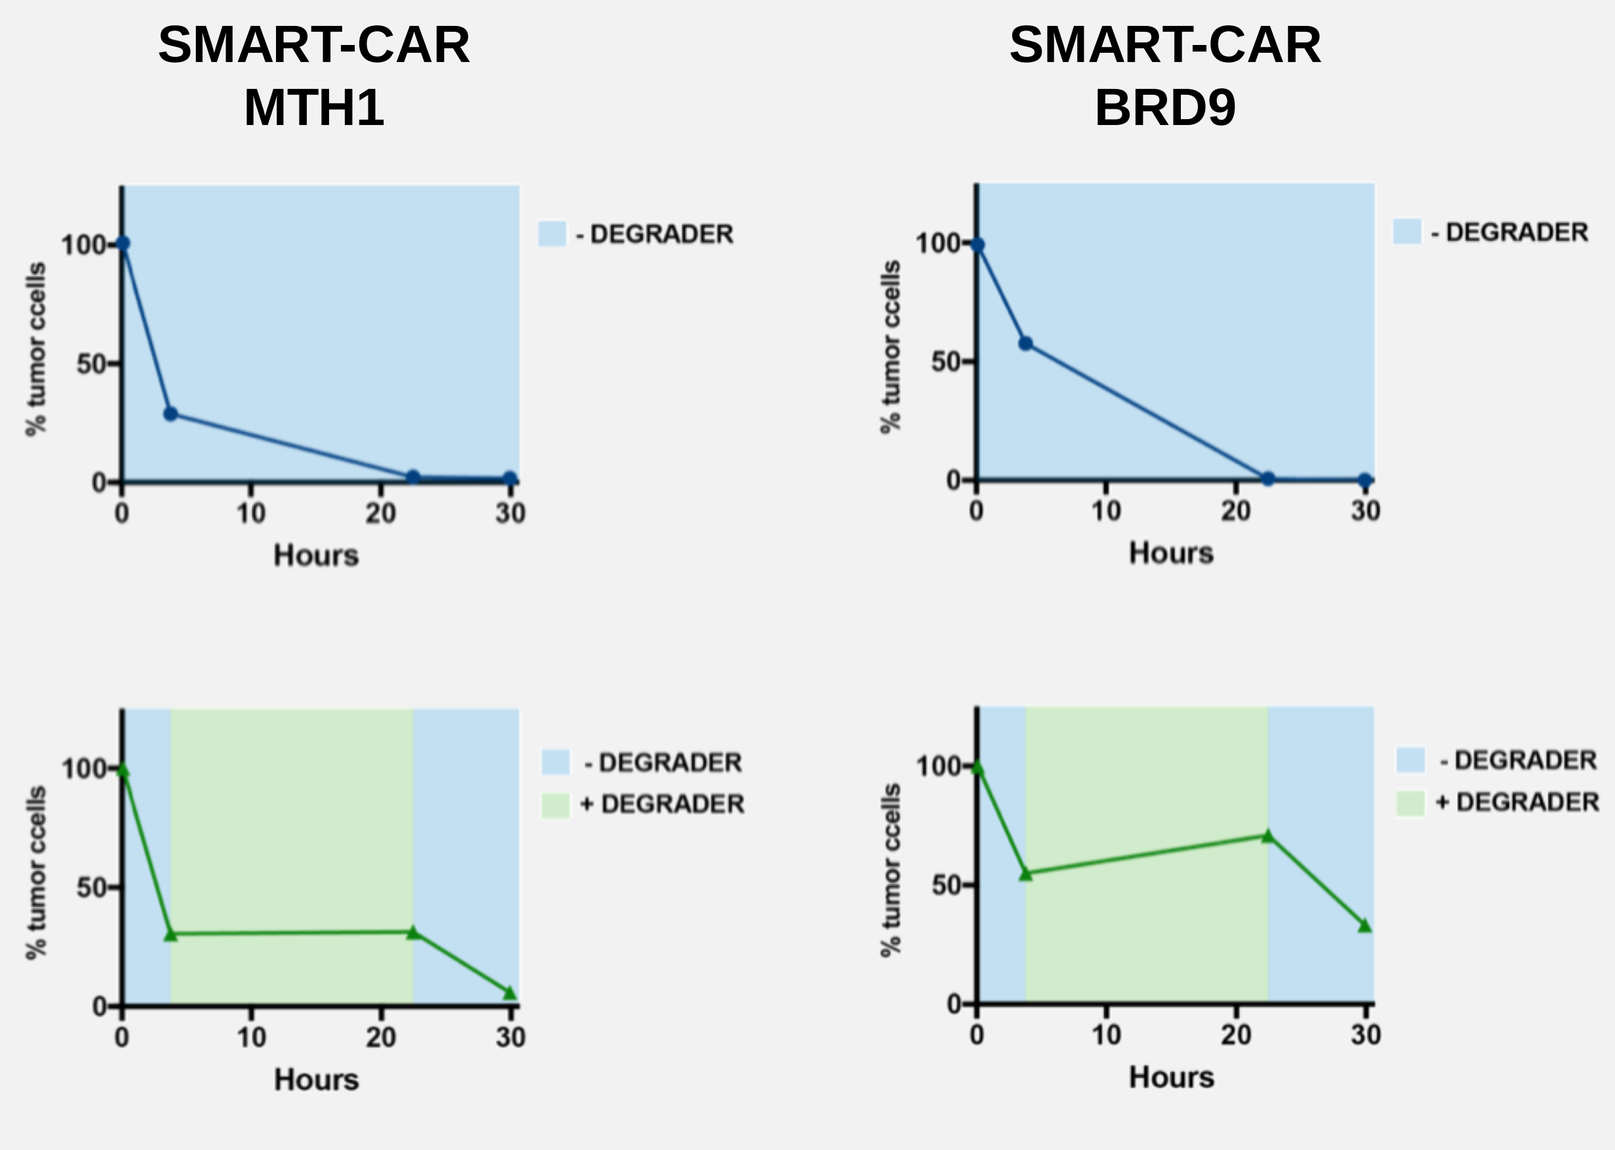
SMART-CAR MTH1
SMART-CAR BRD9
% tumor cells
- DEGRADER
+ DEGRADER
Hours

Functional control of SMART-CAR expressing primary T-cells

Degrader based control of SMART-CAR expressing primary T-cells

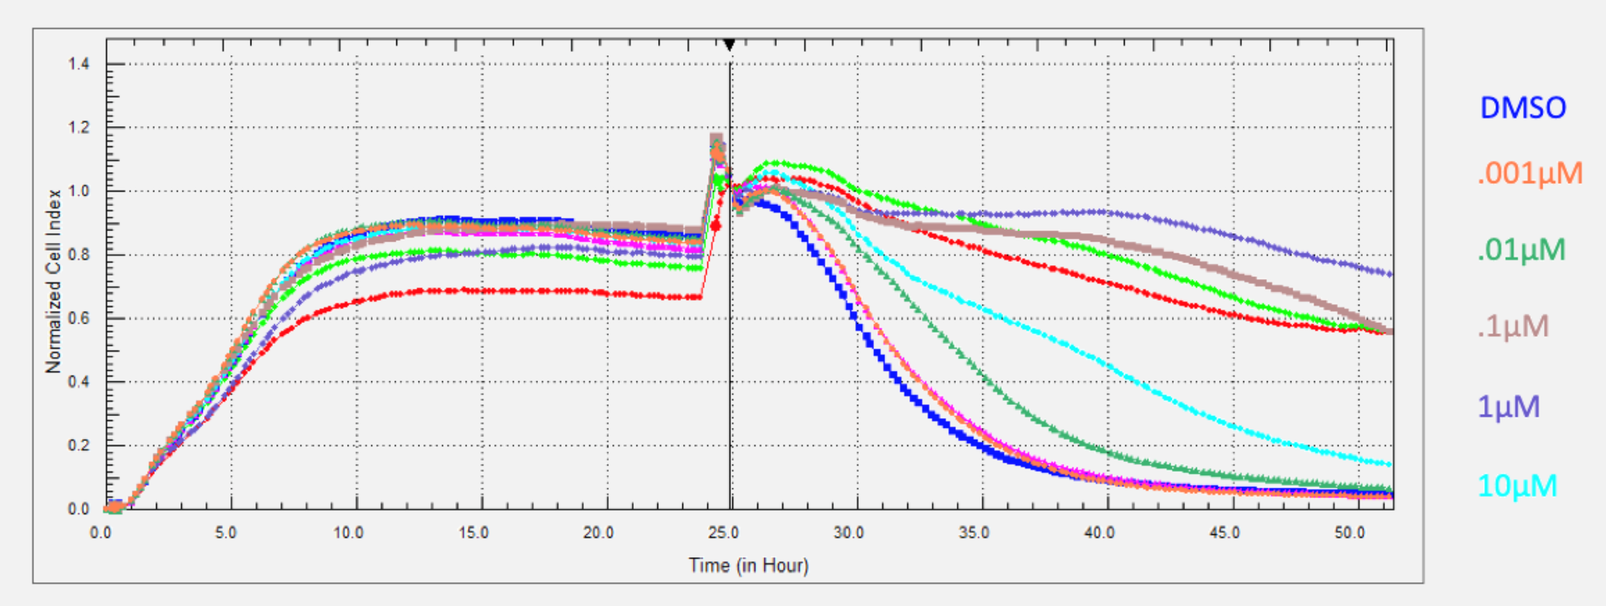
Normalized Cell Index
Time (in Hour)
DMSO
.001µM
.01µM
.1µM
1µM
10µM

Degrader based control of cytokine production

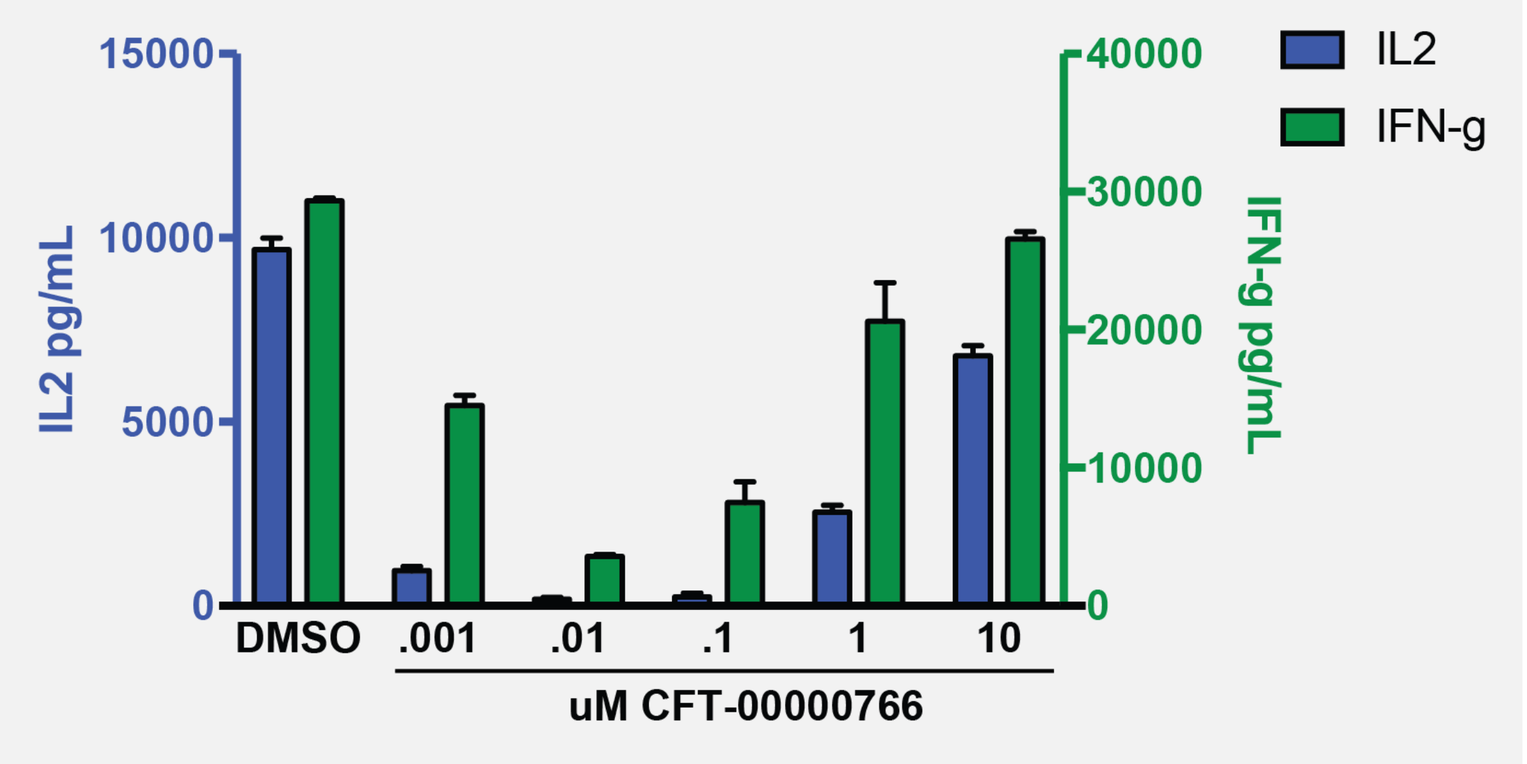
IL2 pg/mL
IFN-g pg/mL
IL2
IFN-g
DMSO
.001
.01
.1
1
10
uM CFT-00000766

OPEN SOURCE

TOOL-KIT

APPLICATIONS

TWO aTAGS + MULTIPLE DEGRADERS
Highly potent and rapid acting
In vitro and *in vivo* applicability

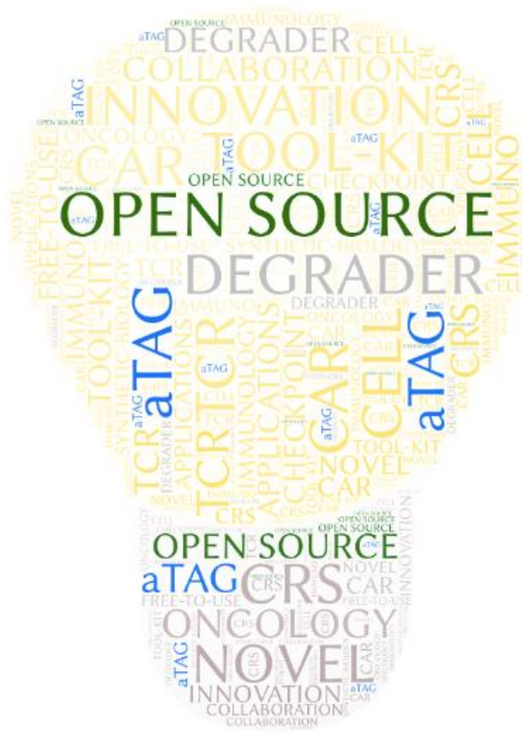
DEGRADER
INNOVATION
TOOL-KIT
OPEN SOURCE
DEGRADER
aTAG
aTAG
OPEN SOURCE
aTAG
CRS
ONCOLOGY
NOVEL

Want to use? E-mail us at:
aTAG@c4therapeutics.com

REFERENCES

1. Nishimura, K., Fukagawa, T., Takisawa, H., Kakimoto, T. & Kanemaki, M. An auxin-based degron system for the rapid depletion of proteins in nonplant cells. Nat Methods 6, 917–922 (2009).
2. Nabet, B. et al. The dTAG system for immediate and target-specific protein degradation. Nat Chem Biol 1–11 (2018). doi:10.1038/s41589-018-0021-8
3. Buckley, D. L. et al. HaloPROTACS: Use of Small Molecule PROTACs to Induce Degradation of HaloTag Fusion Proteins. ACS Chemical Biology 10, 1831–1837 (2015).
4. Neklesa, T. K. et al. Small-molecule hydrophobic tagging-induced degradation of HaloTag fusion proteins. Nature chemical biology 7, 538–43 (2011).
5. Chung, H. K. et al. Tunable and reversible drug control of protein production via a self-excising degron. Nat Chem Biol 11, 713–720 (2015).
6. Bonger, K. M., Chen, L., Liu, C. W. & Wandless, T. J. Small-molecule displacement of a cryptic degron causes conditional protein degradation. Nat Chem Biol 7, 531 (2011).
7. Banaszynski, L. A., Chen, L.-C. C., Maynard-Smith, L. A., Ooi, A. & Wandless, T. J. A rapid, reversible, and tunable method to regulate protein function in living cells using synthetic small molecules. Cell 126, 995–1004 (2006).