MOMENTUM: A Phase 2, Open-Label, Single-Arm, Multicenter Study to Evaluate the Efficacy of Cemsidomide + Dexamethasone in Subjects With Relapsed/Refractory Multiple Myeloma

Binod Dhakal, MD[1], Karthik Ramasamy, MBBS[2], Amro Ali, PharmD[3], Leah Leahy, BS[3], Uma Chandrasekaran, PhD[3], Riadh Lobbardi, PhD[3], Eunju Hurh, PhD[3], Leonard Reyno, MD[3], and Martin Kaiser, MD[4]

[1]Medical College of Wisconsin, Milwaukee, Wisconsin; [2]Oxford University Hospital NHS Trust, Oxford, England, United Kingdom; [3]C4 Therapeutics, Inc., Watertown, MA; [4]Department of Haematology, The Royal Marsden Hospital and The Institute of Cancer Research, London, United Kingdom

BACKGROUND

- Cemsidomide is a novel, highly potent, cereblon-based, IKZF1/3 MonoDAC® degrader, having a similar mechanism of action as BMS's IMiD® or CELMoD® degraders for MM
- Cemsidomide displays catalytic activity enabling rapid and deep target degradation with high binding affinity to overcome resistance due to low cereblon levels
- Cemsidomide binds to cereblon to facilitate the recruitment and ubiquitination of IKZF1/3 leading to the proteasomal degradation of both proteins (Figure 1)
- IKZF1/3 degradation induces multiple myeloma cell death, activation of fully differentiated T-cells which prevents T-cell exhaustion and promotes secretion of key immune stimulating cytokines[1]
- In a Phase 1 study, cemsidomide, given orally QD over 14 days with a 14 day "off" period (14/14) in combination with weekly dexamethasone in a heavily pre-treated patient population (7 median prior lines), was well tolerated and demonstrated durable anti-myeloma activity at increasing dose levels
 - A 53% ORR was observed as the highest dose of 100 µg QD 14/14, with a 36% ORR observed across all dose levels[1,2]

Figure 1: Mechanism of Action of Cemsidomide[3]

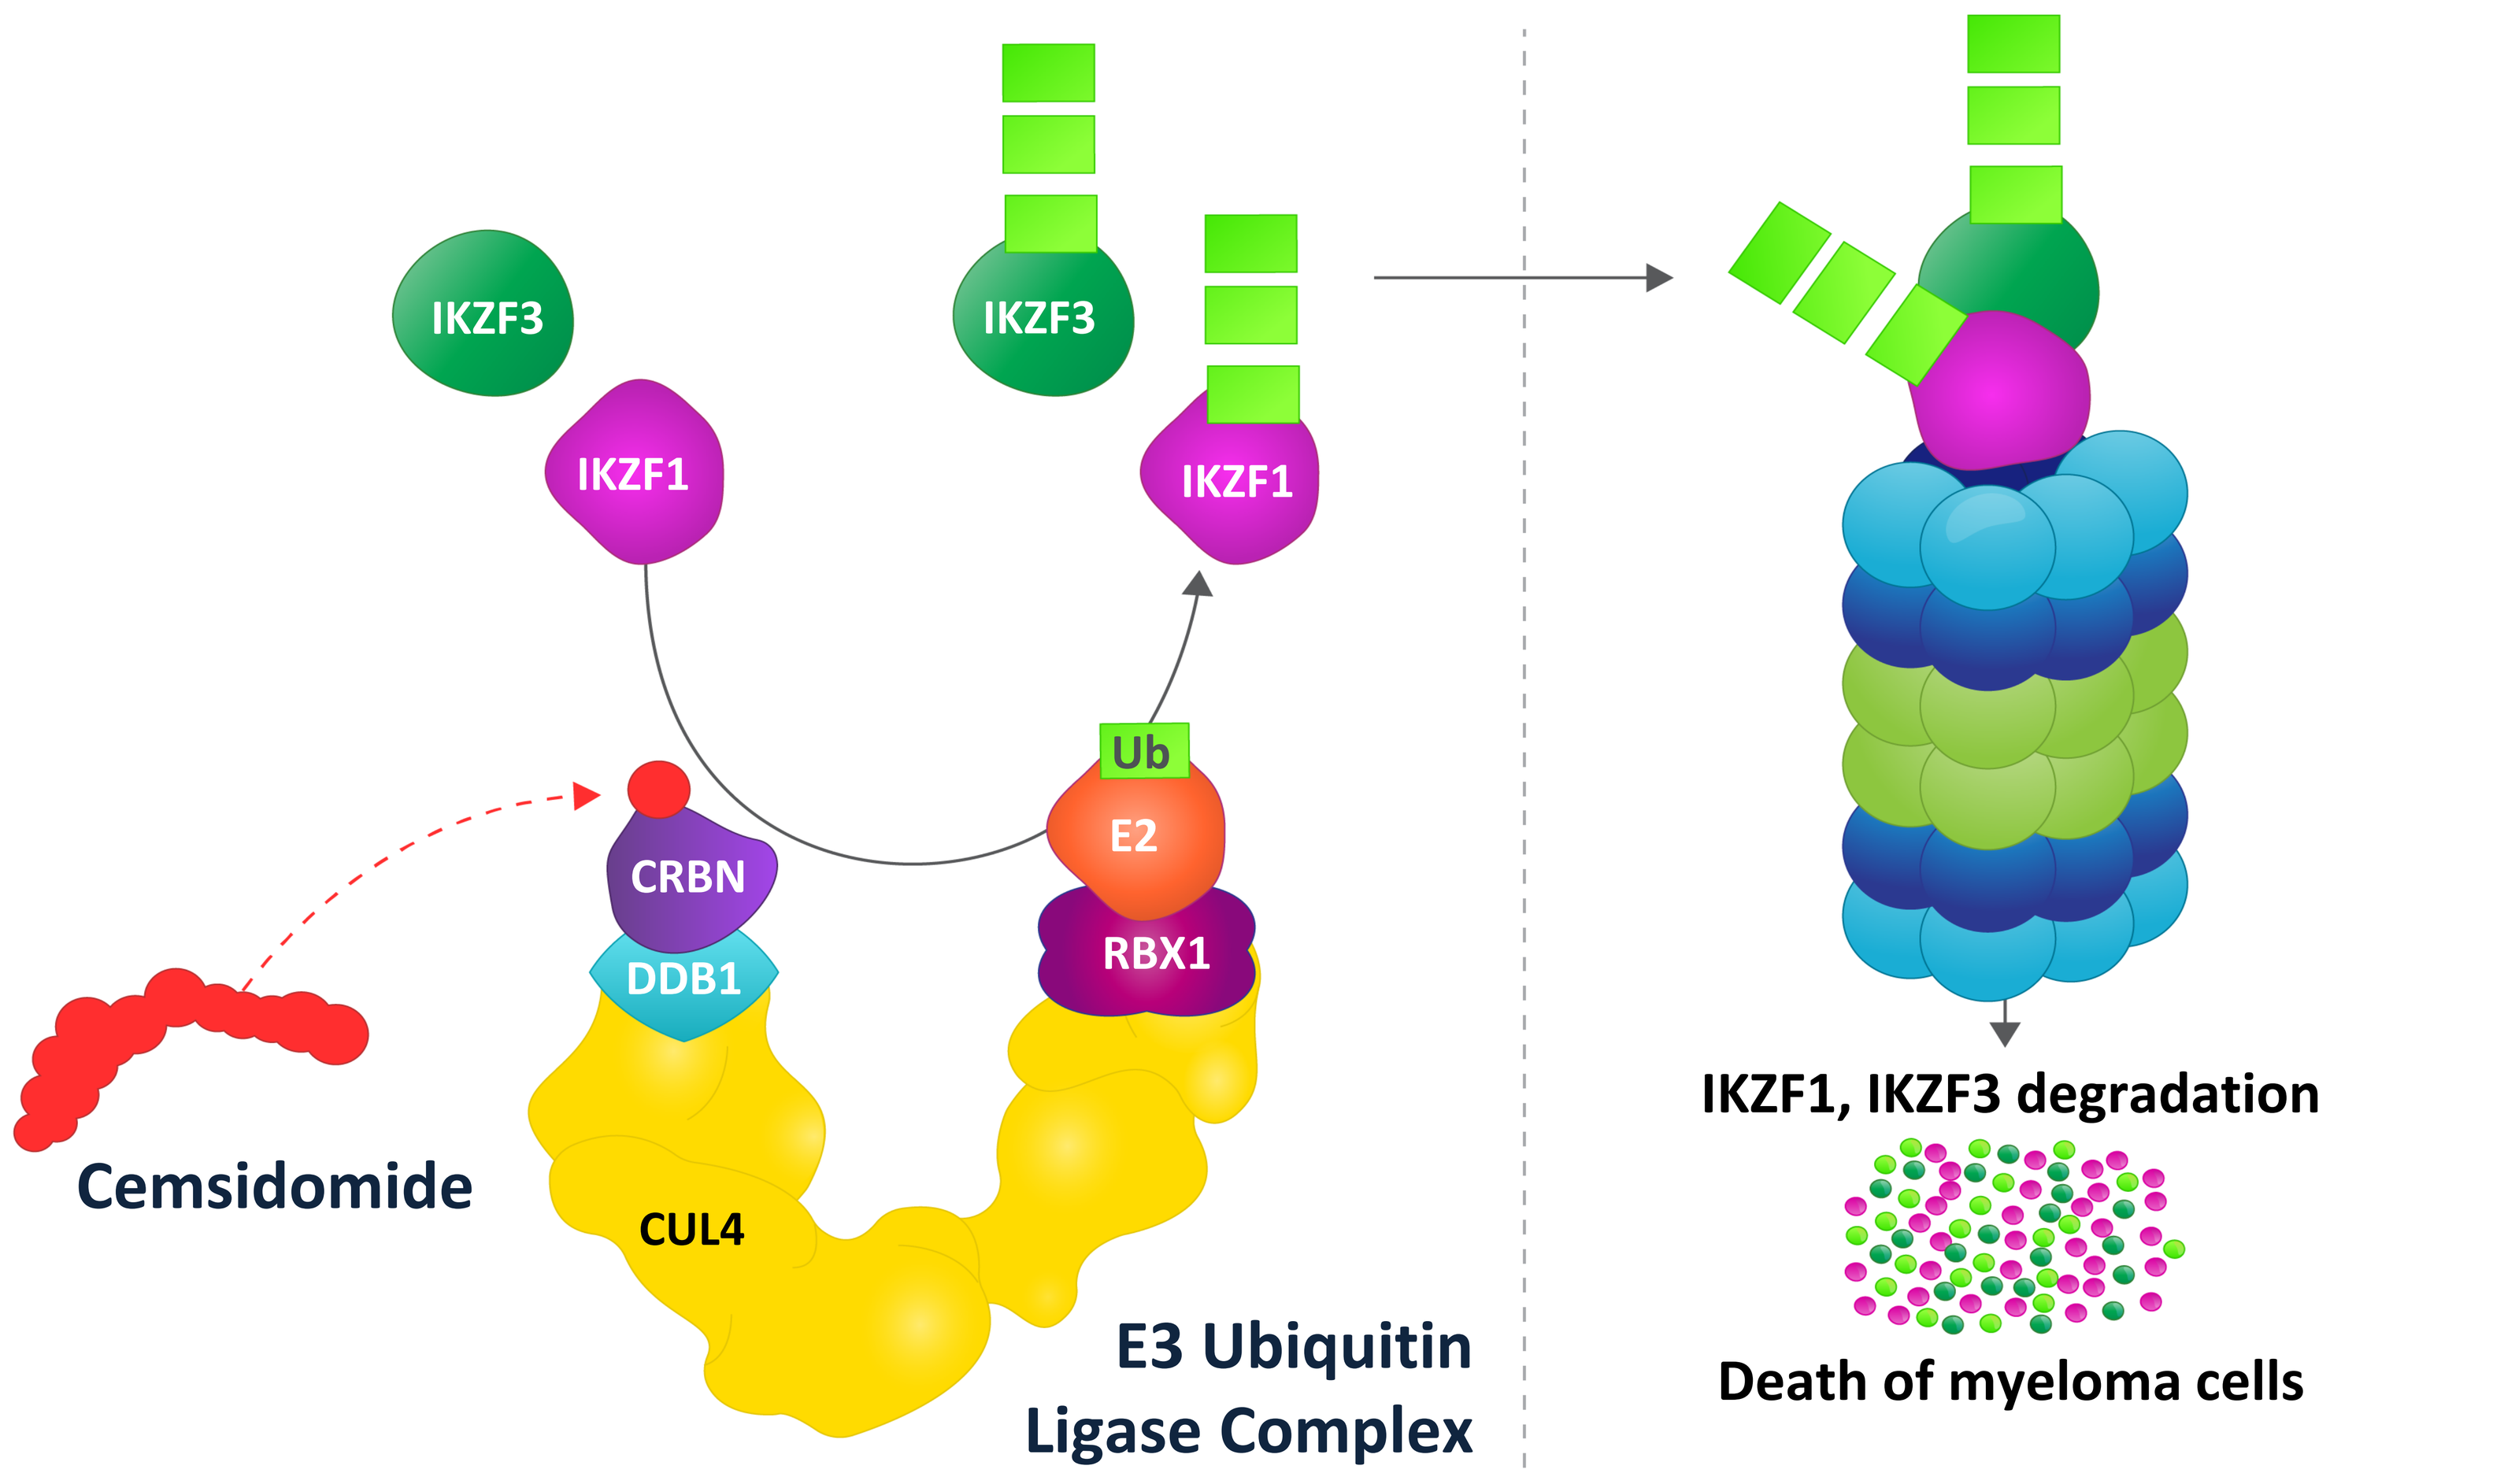
IKZF3
IKZF1
IKZF3
IKZF1
Ub
CRBN
E2
DDB1
RBX1
Cemsidomide
CUL4
E3 Ubiquitin Ligase Complex
IKZF1, IKZF3 degradation
Death of myeloma cells

Objective

- To evaluate the antimyeloma activity and further characterize the safety, tolerability, PK, and PD of cemsidomide in combination with dexamethasone in subjects with relapsed/refractory multiple myeloma

METHODS/STUDY DESIGN

Figure 2: Study Design

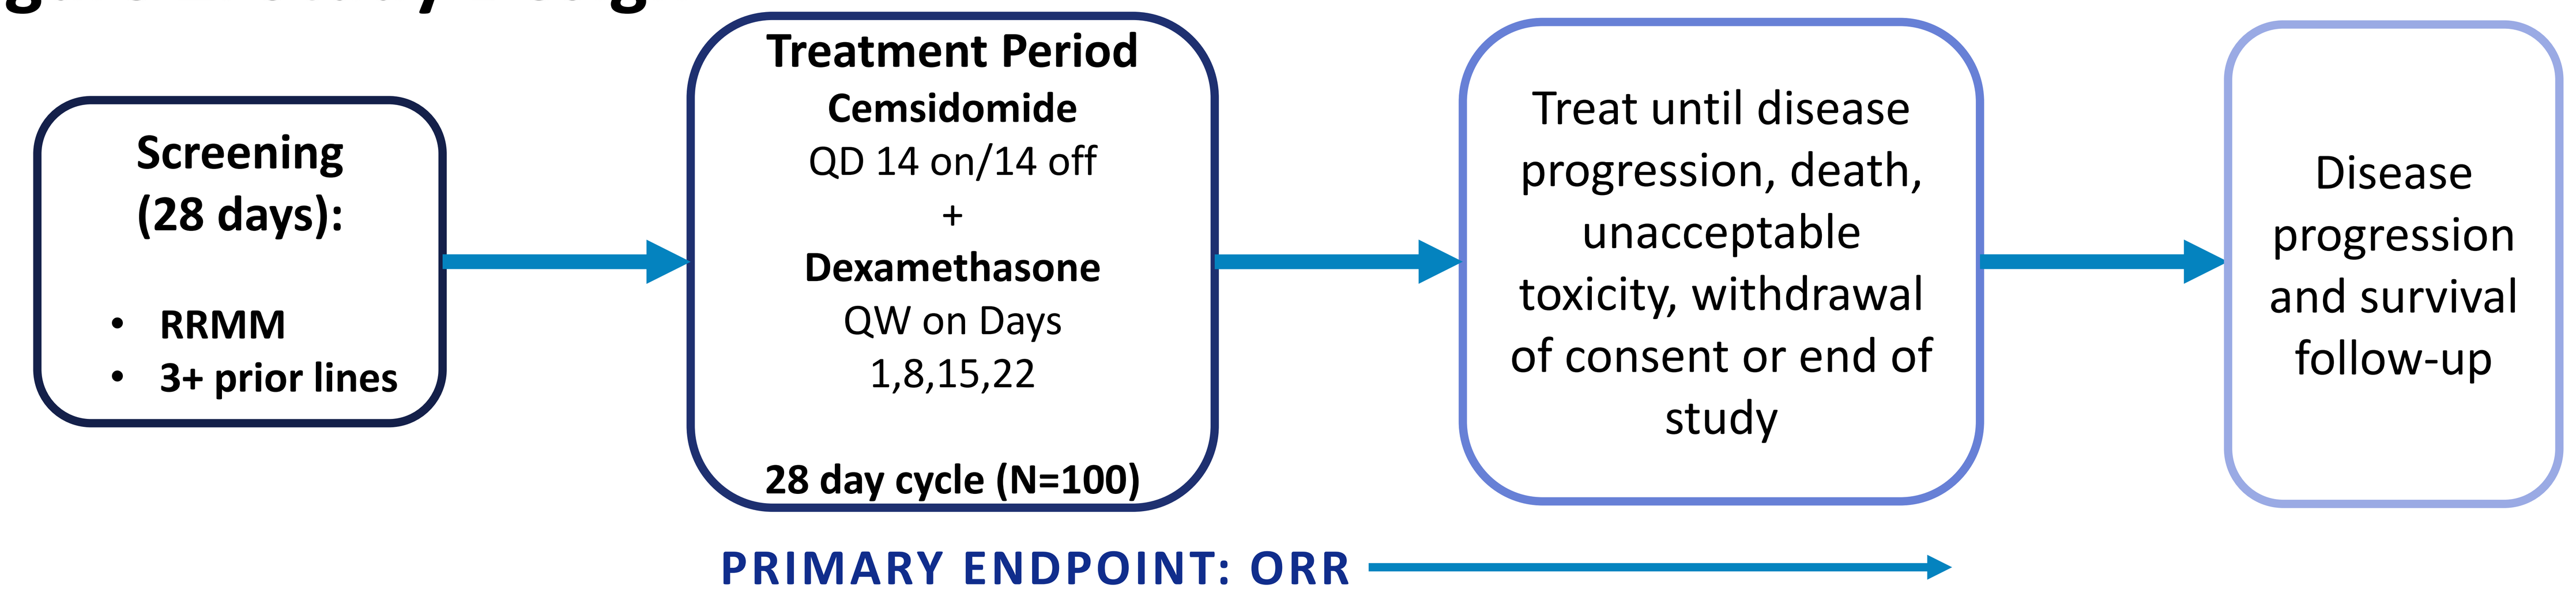
Screening (28 days):
• RRMM
• 3+ prior lines
Treatment Period
Cemsidomide
QD 14 on/14 off
+
Dexamethasone
QW on Days 1,8,15,22
28 day cycle (N=100)
Treat until disease progression, death, unacceptable toxicity, withdrawal of consent or end of study
Disease progression and survival follow-up
PRIMARY ENDPOINT: ORR

KEY ELIGIBILITY CRITERIA

KEY INCLUSION CRITERIA

- ≥18 years of age at time of informed consent
- Subjects must have a documented diagnosis of MM and measurable disease
- Subjects must have received ≥3 prior anti-myeloma regimens including ≥2 consecutive cycles of:
 - Immunomodulatory drug (i.e., IKZF1/3 degrader)
 - Proteasome inhibitor
 - Anti-CD38 antibody
 - TCE or CAR-T therapy (unless not indicated and/or patient refusal)
- Refractory to or had documented disease progression within 60 days from the last dose of their prior MM treatment

KEY EXCLUSION CRITERIA

- Presence of myeloma in the CNS
- Treatment with mezigdomide or iberdomide during the subjects most recent line of therapy
- Nonsecretory or oligosecretory MM

STUDY ENDPOINTS

PRIMARY ENDPOINTS

- ORR per IMWG response criteria by IRC

SECONDARY ENDPOINTS

- DoR (IRC assessed)
- CR rate (include MRD status via local testing; IRC assessed)
- Time to response (IRC assessed)
- PFS (IRC assessed)
- OS
- Safety and tolerability of cemsidomide in combination with dexamethasone
- Plasma concentrations of cemsidomide

EXPLORATORY ENDPOINTS

- ORR (investigator assessed)
- DOR (investigator assessed)
- CR rate (including MRD status via local testing; investigator assessed)
- Time to response (investigator assessed)
- PFS (investigator assessed)
- Degradation of IKZF1/3 in PBMCs
- Change in EORTC QLQ-MY20 Disease Symptoms
- Change in EORTC QLQ-MY20 Treatment Side Effects
- Change in EORTC QLQ-C30 score

STUDY STATUS/ENROLLMENT[4]

The study enrolled its first patient in February 2026 and will be recruiting ~N=100 subjects from 60 sites* in the US and Europe

- Trial registration: NCT07284758[4]
- Contact information: *clinicaltrials@C4therapeutics.com*

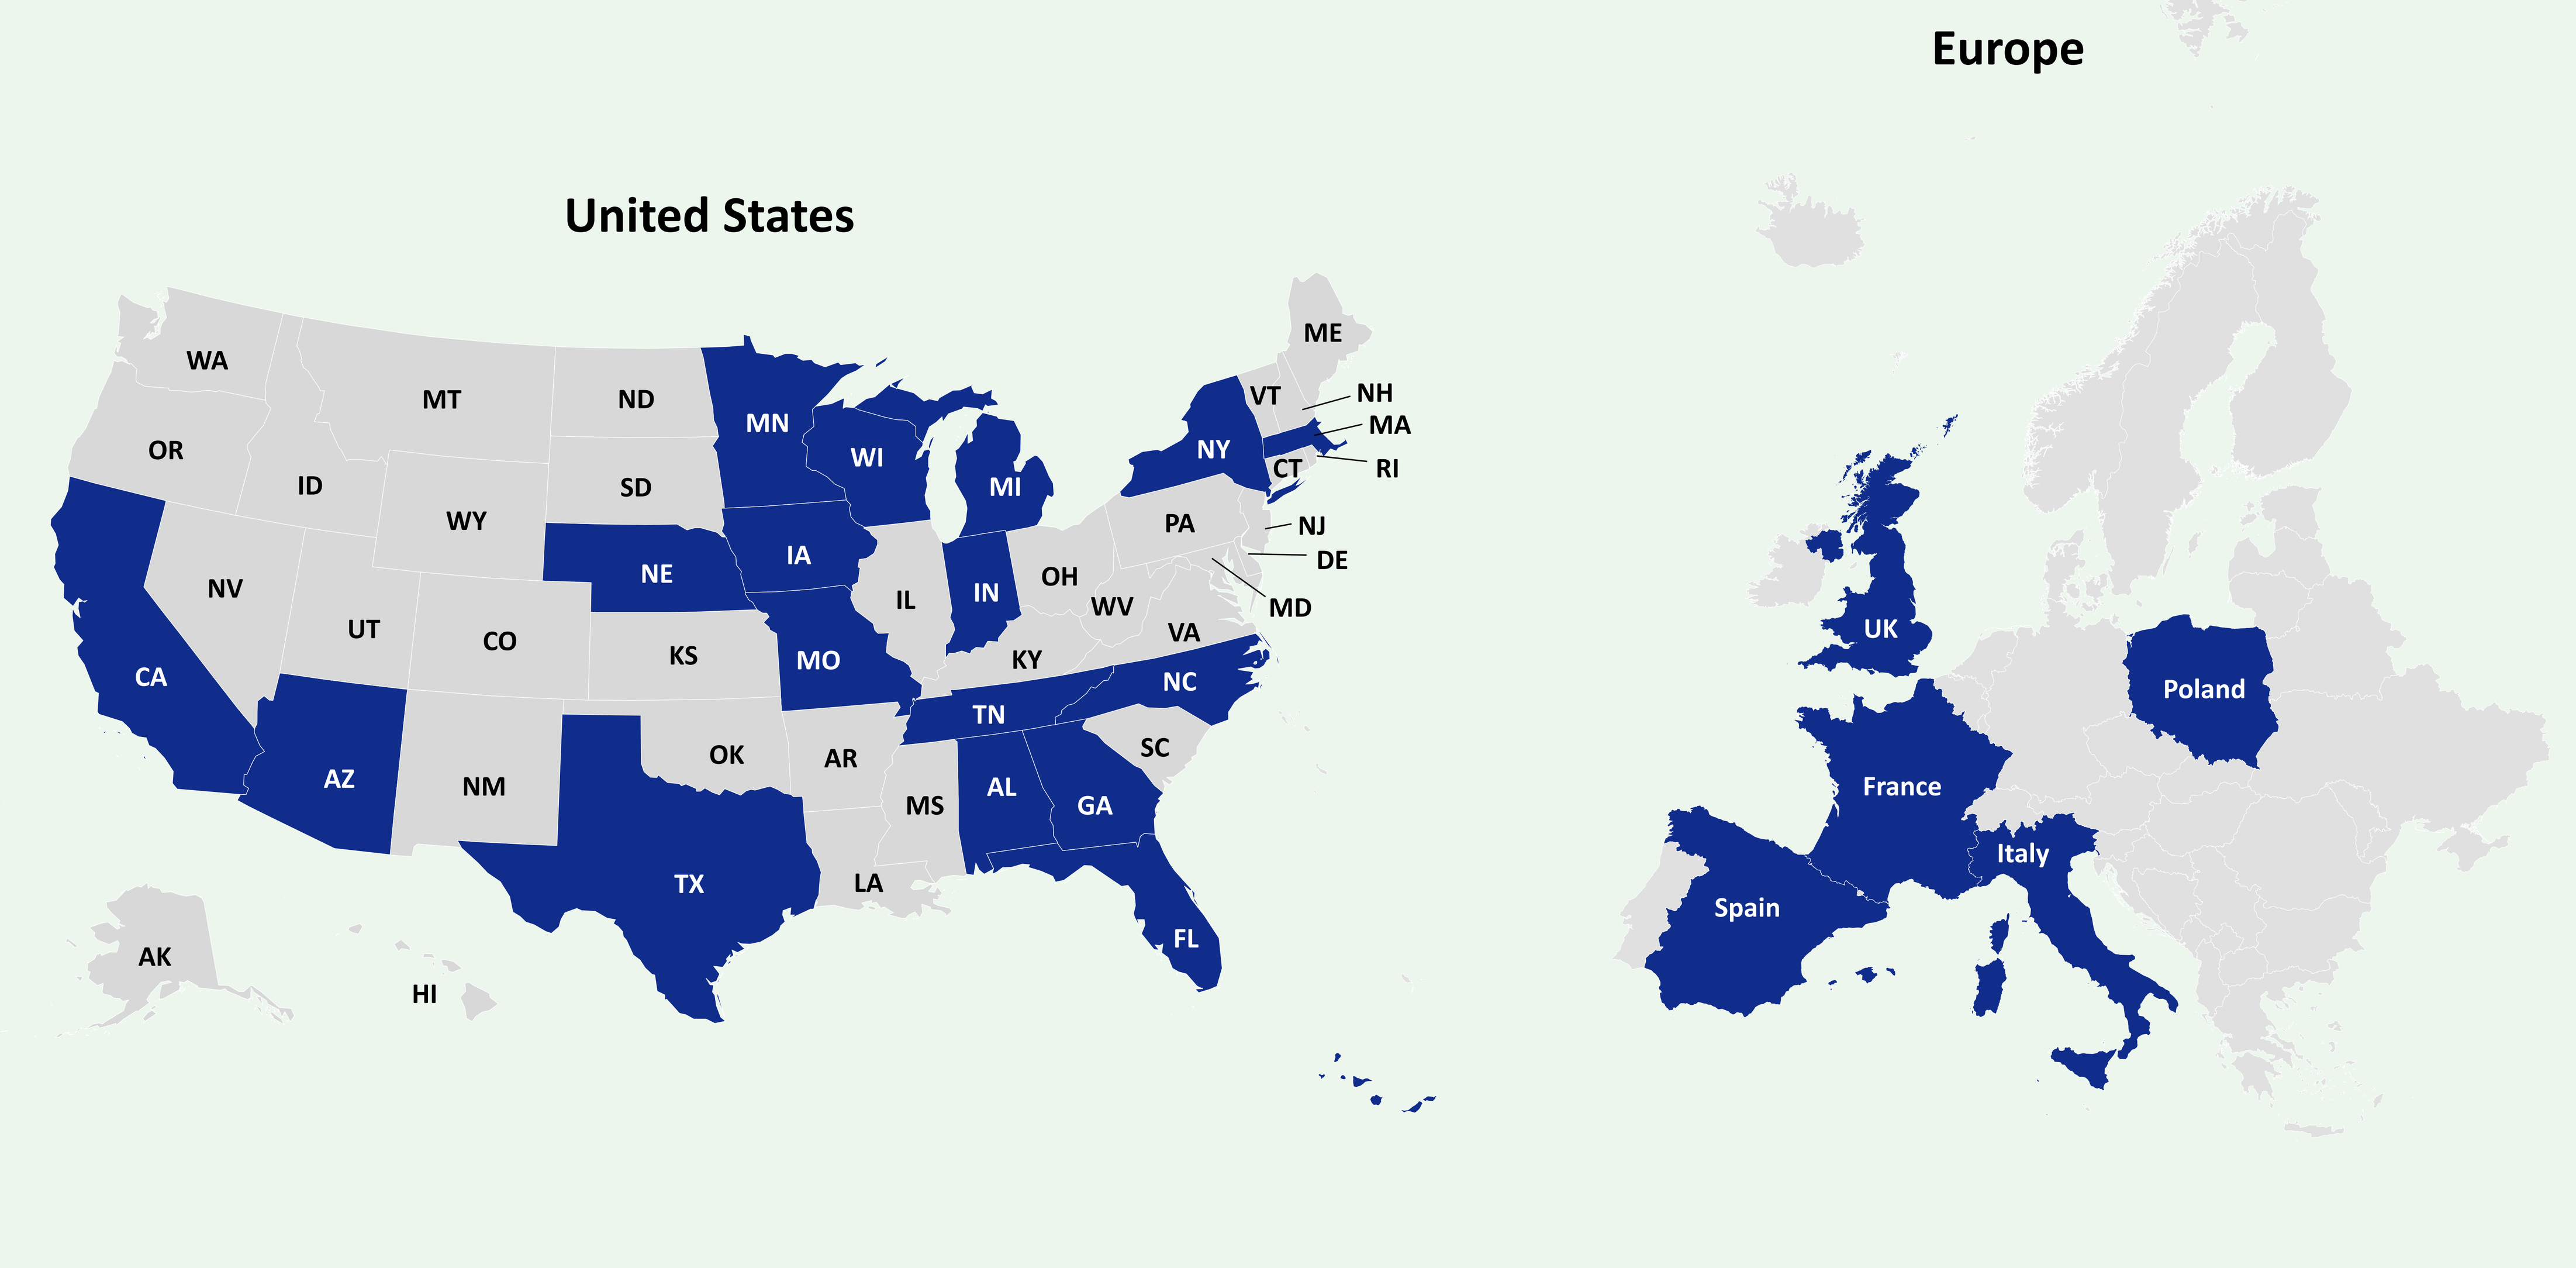
United States
WA
OR
CA
NV
ID
MT
WY
UT
AZ
CO
NM
ND
SD
NE
KS
OK
TX
MN
IA
MO
AR
LA
WI
IL
MS
MI
IN
KY
TN
AL
OH
WV
VA
NC
SC
GA
FL
PA
NY
VT
NH
ME
MA
RI
CT
NJ
DE
MD
AK
HI
Europe
UK
France
Spain
Italy
Poland

*Blue-colored US states and EU countries indicate selected clinical trial sites.

Abbreviations

BMS, Bristol Myers Squibb; **CNS**, Central Nervous System; **CR**, Complete Response; **CRBN**, Cereblon; **DoR**, Duration of Response; **EROTC**, European Organisation for Research and Treatment of Cancer; **IKZF1/3**, Ikaros Family Zinc Finger 1 and 3; **IMWG**, International Myeloma Working Group; **IRC**, Independent Review Committee; **MM**, Multiple Myeloma; **MRD**, Minimal Residual Disease; **ORR**, Overall Response Rate; **OS**, Overall Survival; **PBMCs**, Peripheral Blood Mononuclear Cells; **PD**, Pharmacodynamics; **PFS**, Progression-Free Survival; **PK**, Pharmacokinetics; **QD**, Once daily; **QLQ**, Quality of Life Questionnaire; **QW**, Once weekly; **RRMM**, Relapsed/Refractory Multiple Myeloma; **TCE**, T-Cell Engager; **US**, United States

Disclosures

BD: consulting or advisory role: BMS, Janssen, Arcellx, Sanofi, GSK, Menarini, Genentech, Karyopharm, Pfizer, Kite, AstraZeneca; research funding: BMS, Janssen, Arcellx, Carsgen, Sanofi, Caribou, Gracell, Menarini, C4 Therapeutics; honoraria: Karyopharm, BMS, Janssen. **KR**: consulting or advisory role: Janssen, Adaptive Biotech, Amgen, Recordati, Takeda, AbbVie, Oncopeptides, Celgene – Bristol Myers Squibb, Pfizer, Menarini Stemline, Thermofisher (Binding site), GSK; research funding: Sanofi, Pfizer, GSK, Celgene – Bristol Myers Squibb; honoraria: Janssen, Adaptive Biotech, Amgen, Takeda, Abbvie, Oncopeptides, Celgene – Bristol Myers Squibb, Pfizer, Menarini stemline, Recordati, GSS; travel, accommodations, expenses: BMS, Amgen, Takeda, Recordati. **AA**: employed by and equity holder in C4 Therapeutics. **LL**: employed by and equity holder in C4 Therapeutics. **UC**: employed by and equity holder in C4 Therapeutics. **RL**: employed by and equity holder in C4 Therapeutics. **EH**: employed by and equity holder in C4 Therapeutics. **LR**: employed by and equity holder in C4 Therapeutics. **MK**: consulting or advisory role: AbbVie, Adaptive, BMS/Celgene, Johnson and Johnson, GSK, Karyopharm, Kite, Pfizer, Poolbeg, Regeneron, Roche, Sanofi, Takeda; research funding: BMS/Celgene; honoraria: AbbVie, Adaptive, BMS/Celgene, Johnson and Johnson, GSK, Sanofi, Takeda; travel, accommodations, expenses: BMS/Celgene, Johnson and Johnson, Takeda.

Acknowledgements

We would like to thank the site support staff, study sponsor, and collaborators as well as participating patients and their families for their contributions to the study. This study is sponsored by C4 Therapeutics, Inc. All authors contributed to and approved the presentation.

References

1. Dhakal B, et al. *Clinical Lymphoma, Myeloma & Leukemia*, Vol. 25, No. S2, S35, 2025
2. C4 Therapeutics data on file.
3. Henderson J, et al. *Cancer Res* (2022) 82 (12_Supplement): ND13
4. NCT07284758. www.clinicaltrials.gov. Accessed April 30, 2026.

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from ASCO and the author.

Presented at The American Society of Clinical Oncology Annual Meeting (ASCO 2026), May 29 - June 2